Small Cell Lung Cancer and Mental Health: European Experience From Lung Cancer Europe's 10th Report

Lung Cancer Europe

Authors: D. Villalón, A. Paraskevopoulou-Souri, A. Brutti, L. Magee, M. Hennink, N. Ehrhardt, D. Montague

INTRODUCTION

The diagnosis of lung cancer extends far beyond the physical. Lung Cancer Europe is an independent non-profit organisation advocating for equitable, high-quality care for people affected by lung cancer. Lung Cancer Europe's 10th Annual Report explored the psychological impact of lung cancer. Extrapolation of Small Cell Lung Cancer (SCLC) data forms part of the wider 11th report.

METHODS

A structured literature review informed survey development. A **46-item** (43 closed; 3 optional open questions) confidential, self completion questionnaire was designed and translated into **19 languages**. Most questions were optional; only filter questions required a response. Dissemination was via Lung Cancer Europe and member organisation channels 28/05/2025-06/07/2025.

This resulted in the **largest European survey on the mental health of people impacted by lung cancer**. Analyses were conducted using SurveyMonkey® tools and Jamovi® software. The SCLC Report relates to the responses of **140** SCLC participants (**6.4%**) out of the total **2,204** respondents.

CONCLUSION

The emotional impact of lung cancer deeply disrupts the daily lives of people living with the disease and those in a caregiving capacity.

Healthcare systems and policy makers must recognise mental health support as a fundamental part of the lung cancer care pathway. Healthcare professionals must provide clear SCLC information and empower individuals to manage their disease and mental well-being.

RESULTS

76 (54.3%) of participants were people diagnosed with SCLC, **64 (45.7%)** caregivers; **111 (79.3%)** female. **66.2%** reported a negative impact of SCLC on their mental health, compared to the overall average of **61.2%**. Fear that the treatment may not work **(72.7%)** and sadness regarding the impact on loved ones **(48.9%)** were the most cited emotional difficulties. **59%** of respondents reported not being offered any form of mental health support. Views regarding seeking mental health are highlighted in Figure 1. The most commonly reported unmet needs are represented in Figure 2.

SURVEY COMPLETION:

76

PARTICIPANTS (OR 54.3%) WERE DIAGNOSED WITH SCLC

66.2%

REPORTED A NEGATIVE IMPACT ON THEIR MENTAL HEALTH DUE TO FEAR OR SADNESS

59%

REPORTED NOT OFFERED ANY MENTAL HEALTH SUPPORT

Figure 1: Have you felt reluctant to seek mental health support following the lung cancer diagnosis for any of the following reasons?

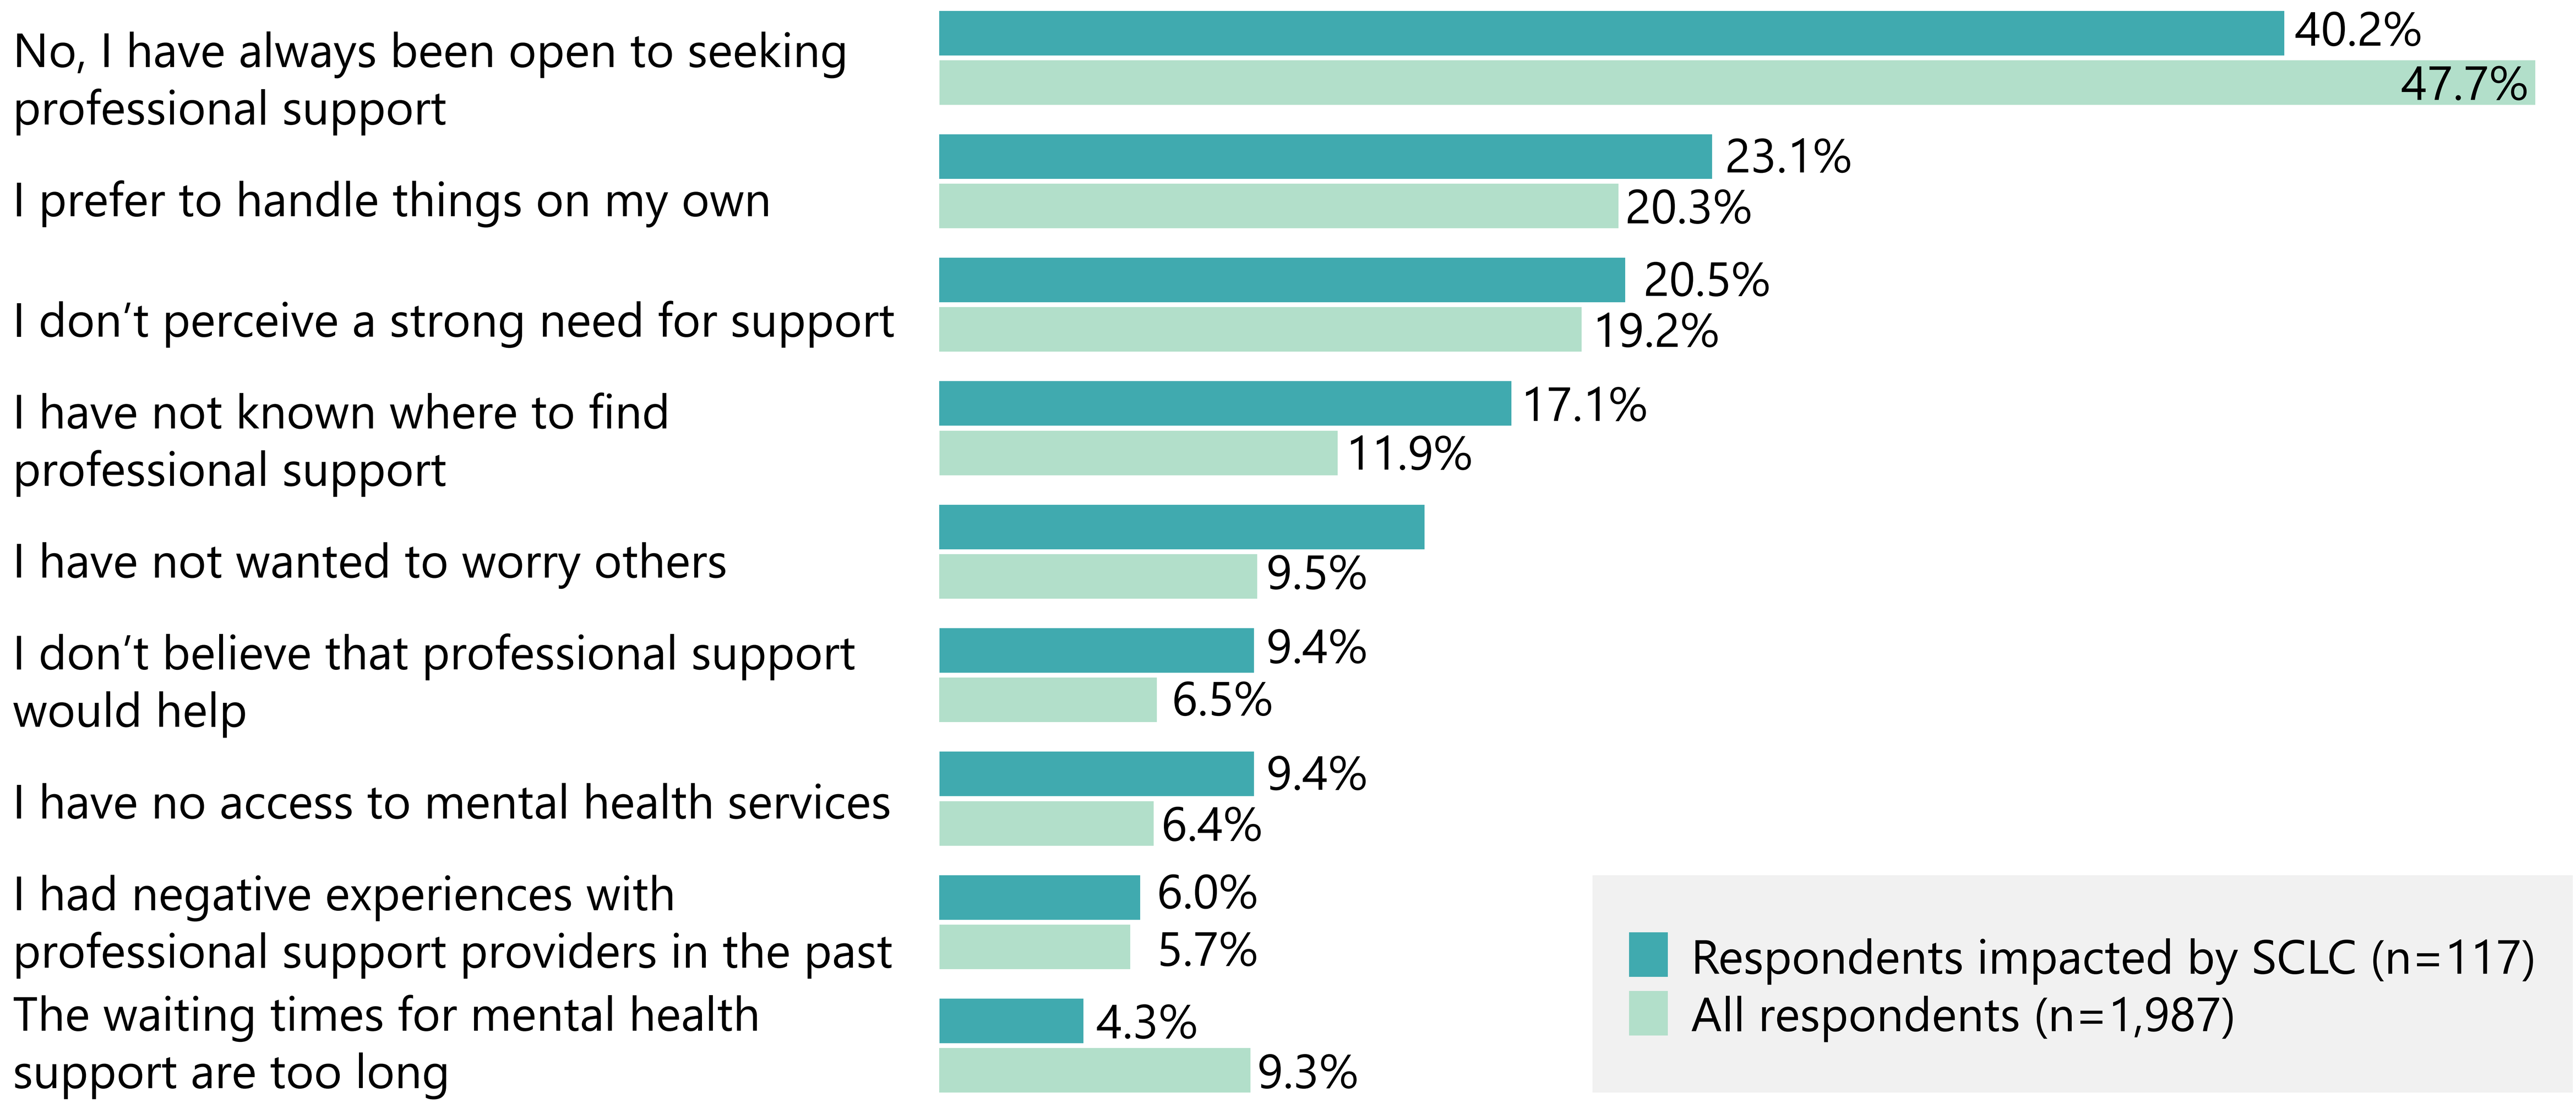
No, I have always been open to seeking professional support
40.2%
47.7%
I prefer to handle things on my own
23.1%
20.3%
I don't perceive a strong need for support
20.5%
19.2%
I have not known where to find professional support
17.1%
11.9%
I have not wanted to worry others
9.5%
I don't believe that professional support would help
9.4%
6.5%
I have no access to mental health services
9.4%
6.4%
I had negative experiences with professional support providers in the past
6.0%
5.7%
The waiting times for mental health support are too long
4.3%
9.3%
Respondents impacted by SCLC (n=117)
All respondents (n=1,987)

Figure 2: Which of the following measures do you think could help improve your mental health?

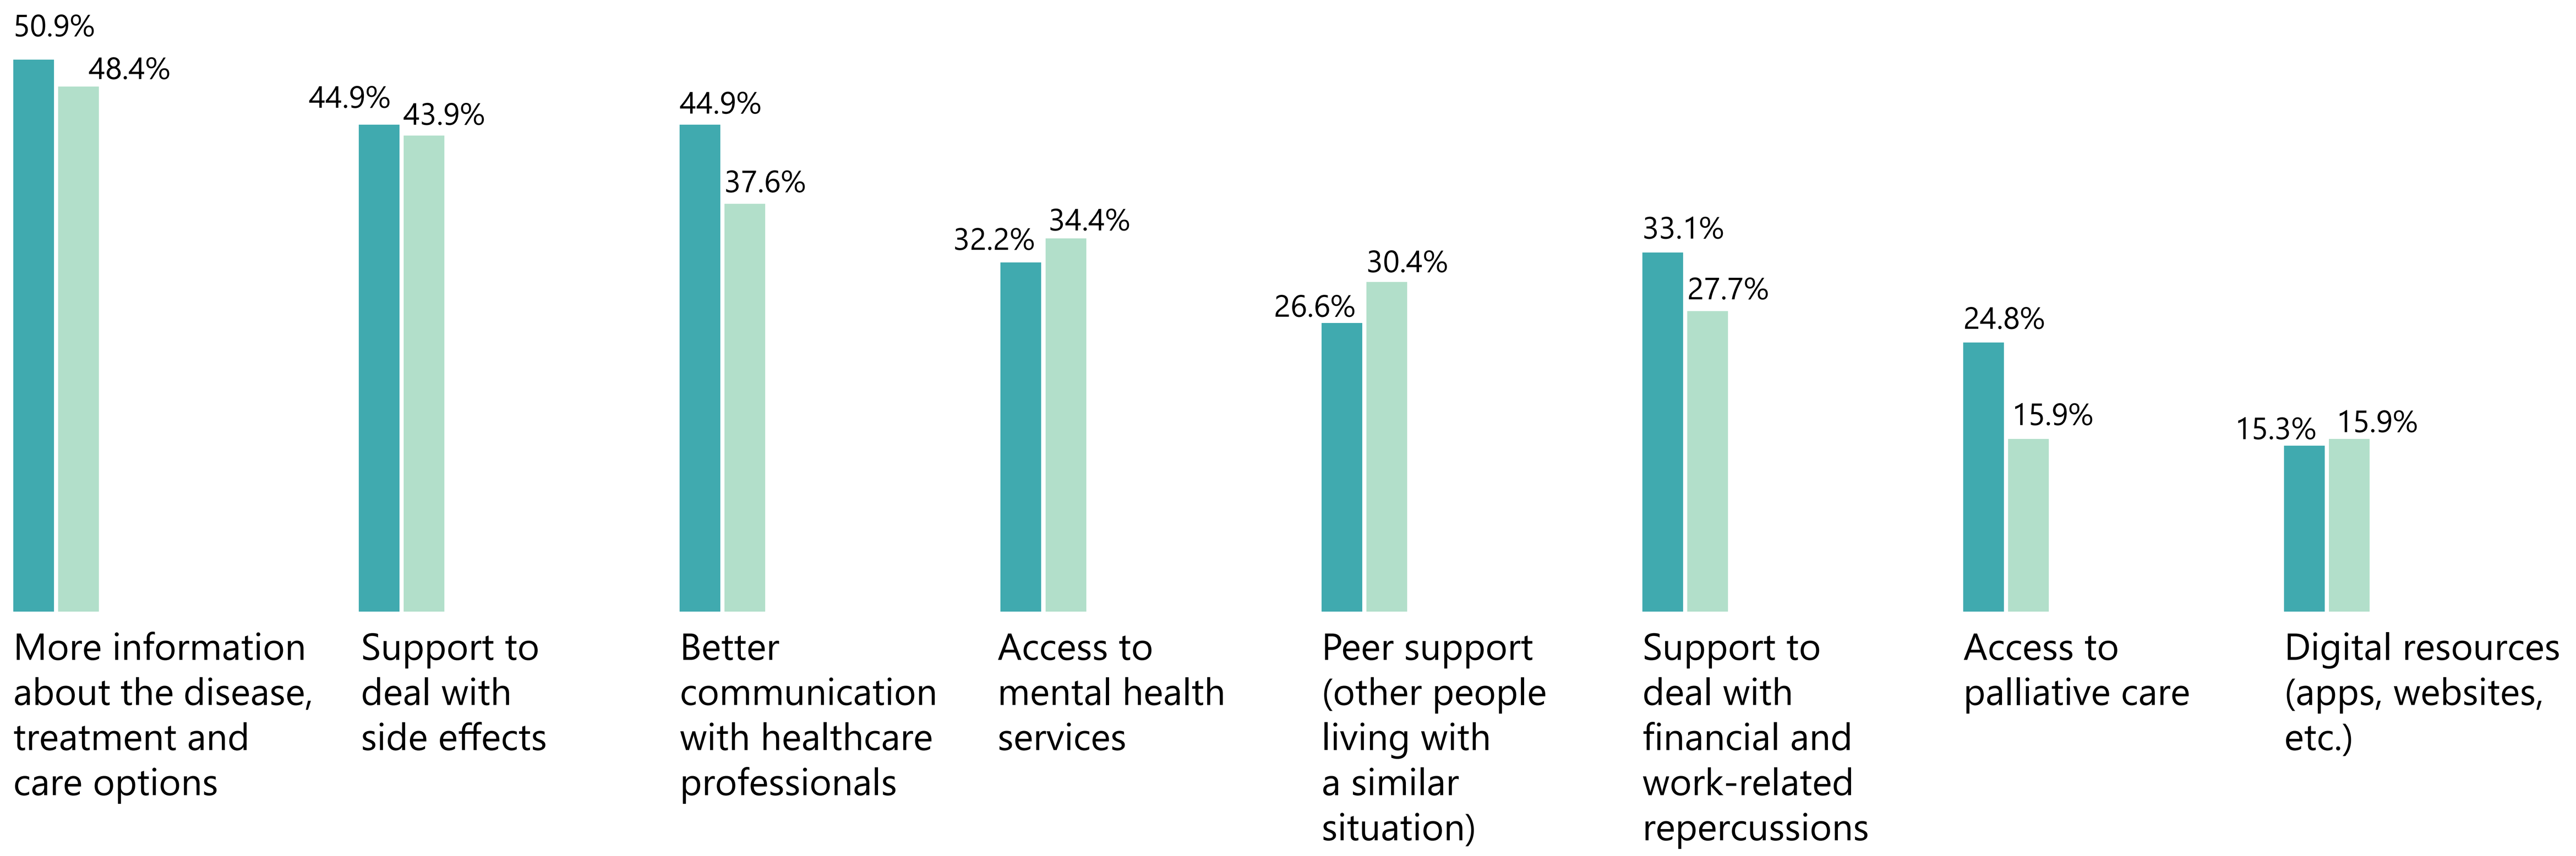
50.9%
48.4%
More information about the disease, treatment and care options
44.9%
43.9%
Support to deal with side effects
44.9%
37.6%
Better communication with healthcare professionals
32.2%
34.4%
Access to mental health services
26.6%
30.4%
Peer support (other people living with a similar situation)
33.1%
27.7%
Support to deal with financial and work-related repercussions
24.8%
15.9%
Access to palliative care
15.3%
15.9%
Digital resources (apps, websites, etc.)